SITC 2023
Abstract Number: 491

Generation of an innovative BsAb targeting CCR8/CTLA4

Qinglin Du, Tiantian Zhou, Yiqi Cao, Fei Peng, Xueyan Yang, Fei Hu, Qian Ding, Yongcong Tan, Shuhua Han
Genor Biopharma Co. Ltd, Shanghai, China

Introduction

CTLA-4-expressing regulatory T cells (Tregs) are abundantly present in tumor tissues and suppress anti-tumor responses. Currently approved CTLA-4 inhibitors, such as ipilimumab, induce severe irAEs, which limited their use. Therefore, next-generation of CTLA-4 inhibitors with reduced toxicity and increased efficacy is highly demanded, given the fact that CTLA-4 inhibition has showed unique advantages in inducing immune memory response and prolonging anti-tumor activity.

CCR8 is recently found to be predominantly expressed on intra-tumor Tregs but little on peripheral Tregs or activated Tconv. Anti-CCR8 antibodies have been shown significant anti-tumor activity in pre-clinical studies. We have hypothesized that a strong anti-CCR8 arm may provide preferential inhibition of CTLA-4 on CCR8+CTLA-4+ intra-tumor Tregs and have synergistic effects on Tregs in TME. Therefore, we have developed a novel BsAb, GBD201, targeting CCR8/CTLA-4. The innovative bsAb exhibited potent depletion of intra-tumor Tregs but spared peripheral Tregs and Tconv. In hCCR8/hCTLA-4 mice, GBD201 showed better efficacy and safety profile compared to ipilimumab.

Molecular structure and mechanisms of GBD201

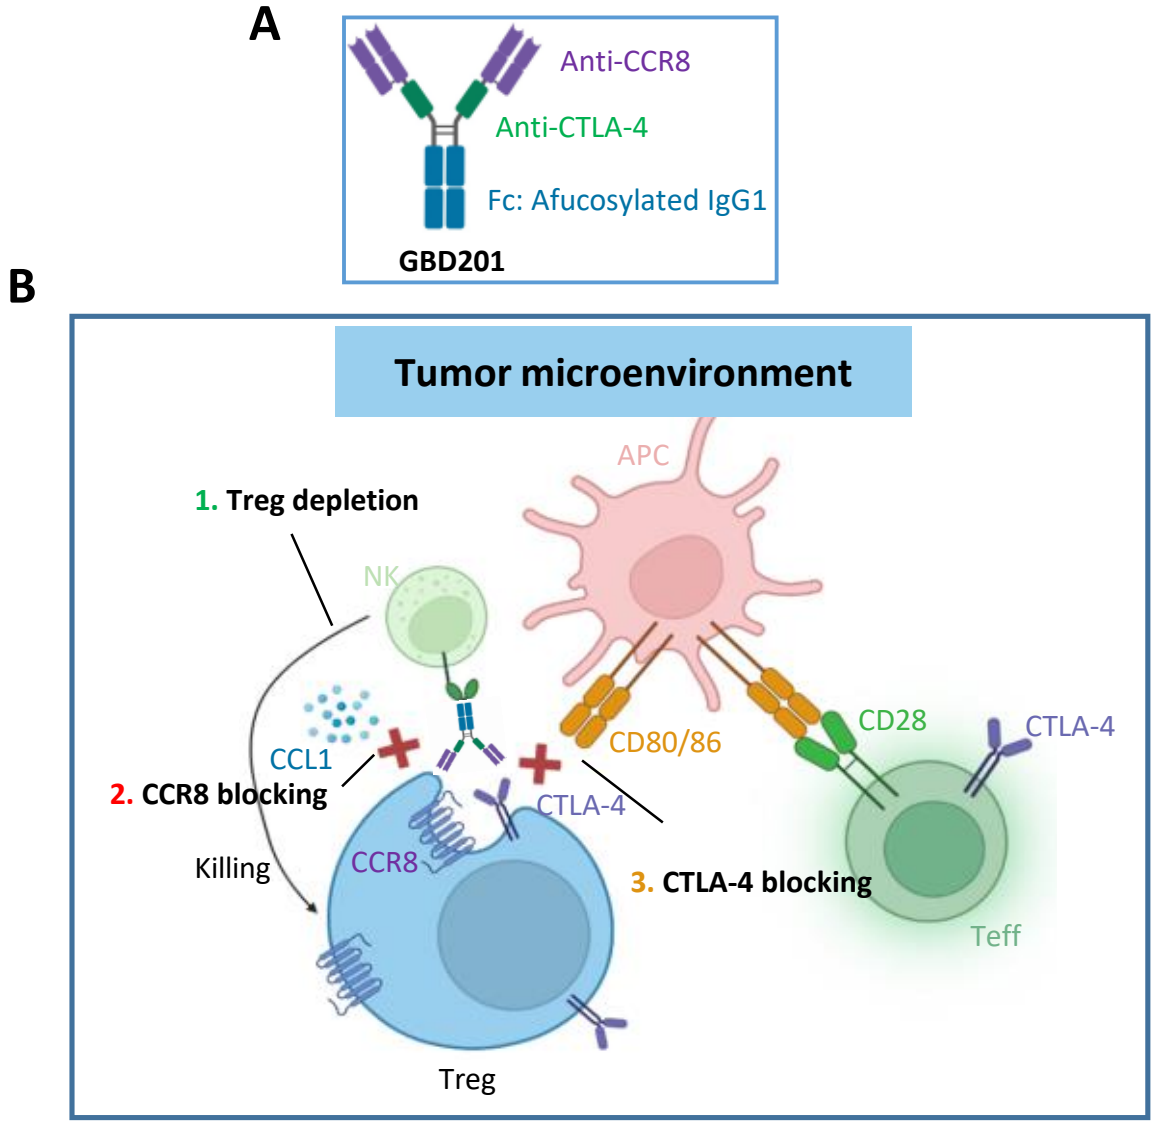
A
Anti-CCR8
Anti-CTLA-4
Fc: Afucosylated IgG1
GBD201
B
Tumor microenvironment
APC
1. Treg depletion
NK
CCL1
2. CCR8 blocking
CD80/86
CD28
CTLA-4
CTLA-4
Killing
CCR8
3. CTLA-4 blocking
Teff
Treg

Figure 1. Molecular structure and multiple mechanisms of GBD201. A. The schematic structure of GBD201. B. The MOAs of GBD201 include the followings: 1) Preferential Treg depletion in TME by specifically binding to CCR8+CTLA-4+ Tregs and enhancing ADCC function. 2) Inhibition of CCR8-CCL1 interaction to block Treg migration and differentiation in TME. 3) Partially blockade of CTLA-4 binding to its ligands CD80/CD86, which is dependent on CCR8 expression.

CCR8 & CTLA-4 are co-expressed on tumor Tregs

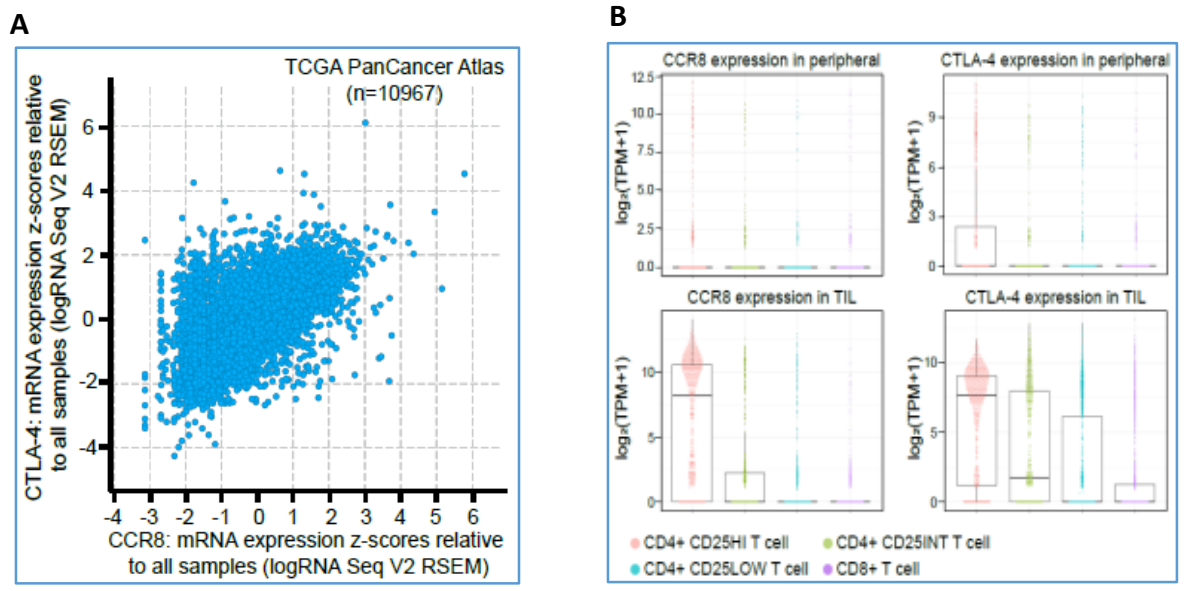
A
TCGA PanCancer Atlas
(n=10967)
CTLA-4: mRNA expression z-scores relative to all samples (logRNA Seq V2 RSEM)
CCR8: mRNA expression z-scores relative to all samples (logRNA Seq V2 RSEM)
B
CCR8 expression in peripheral
CTLA-4 expression in peripheral
CCR8 expression in TIL
CTLA-4 expression in TIL
log2(TPM+1)
CD4+ CD25HI T cell
CD4+ CD25INT T cell
CD4+ CD25LOW T cell
CD8+ T cell

Figure 2. CCR8 and CTLA-4 are highly co-expressed on tumor Tregs. A) Correlation analysis of CCR8 and CTLA-4 mRNA expression levels in tumors from TCGA dataset using cBioportal. **B)** Expression of CCR8 and CTLA-4 in T cells of NSCLC patients by single-cell sequencing according to literature (Xinyi Guo *et al., Nature Medicine*, 2018).

Table 1. Ag-binding affinity measured by Octet

Antigen	Antibody	KD (M)	Kon (1/Ms)	Kdis (1/s)
CCR8 (Met1-Lys35)-mFc	BMK1*	3.436E-09	1.371E05	4.711E-04
	Parental Anti-CCR8-AF**	2.612E-09	1.547E05	4.040E-04
	GBD201	1.592E-09	1.556E05	2.476E-04
CTLA-4-mFc	Ipilimumab	1.339E-09	2.674E05	3.582E-04
	Parental Anti-CTLA-4	8.075E-10	2.277E05	1.839E-04
	GBD201	9.868E-10	3.072E05	3.032E-04

** BMK1 is an afucosylated anti-CCR8 mAb currently in clinical stage.*
***AF: afucosylated*

Differential binding property of GBD201 to CCR8/CTLA-4 double-positive cells vs. CTLA-4 single-positive cells

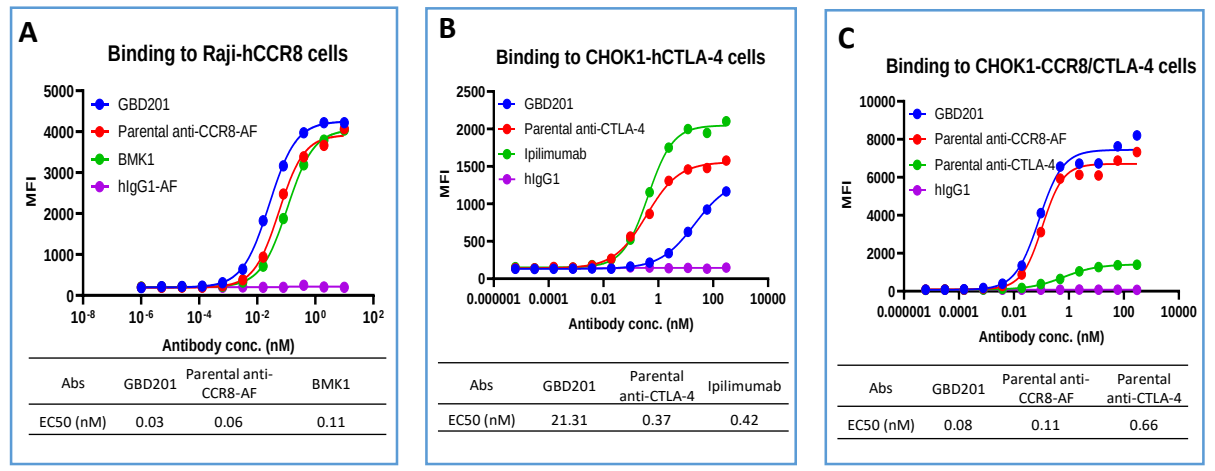
A
Binding to Raji-hCCR8 cells
GBD201
Parental anti-CCR8-AF
BMK1
hIgG1-AF
MFI
Antibody conc. (nM)
Abs | GBD201 | Parental anti-CCR8-AF | BMK1
EC50 (nM) | 0.03 | 0.06 | 0.11
B
Binding to CHOK1-hCTLA-4 cells
GBD201
Parental anti-CTLA-4
Ipilimumab
hIgG1
Abs | GBD201 | Parental anti-CTLA-4 | Ipilimumab
EC50 (nM) | 21.31 | 0.37 | 0.42
C
Binding to CHOK1-CCR8/CTLA-4 cells
GBD201
Parental anti-CCR8-AF
Parental anti-CTLA-4
hIgG1
Abs | GBD201 | Parental anti-CCR8-AF | Parental anti-CTLA-4
EC50 (nM) | 0.08 | 0.11 | 0.66

Figure 3. GBD201 preferentially bound to CCR8-expressing cells. A. GBD201 bound strongly to hCCR8-expressing cells (EC50 = 0.03 nM). **B.** GBD201 relatively weakly bound to hCTLA-4-expressing cells (EC50 = 21.31 nM). **C.** GBD201 potently bound to hCCR8/hCTLA-4 double positive cells (EC50 = 0.08 nM).
BMK1 is a clinical asset of afucosylated anti-CCR8 mAb.

GBD201 efficiently blocked the CCL1/CCR8 interaction

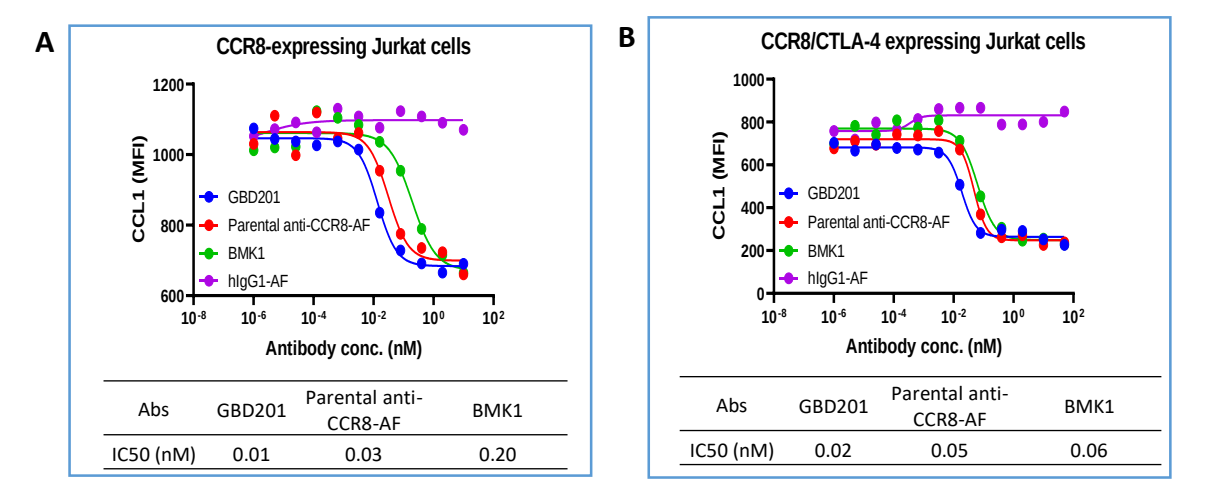
A
CCR8-expressing Jurkat cells
CCL1 (MFI)
GBD201
Parental anti-CCR8-AF
BMK1
hIgG1-AF
Antibody conc. (nM)
Abs | GBD201 | Parental anti-CCR8-AF | BMK1
IC50 (nM) | 0.01 | 0.03 | 0.20
B
CCR8/CTLA-4 expressing Jurkat cells
Abs | GBD201 | Parental anti-CCR8-AF | BMK1
IC50 (nM) | 0.02 | 0.05 | 0.06

Figure 4. GBD201 significantly inhibited the binding of CCL1 to CCR8. GBD201 blocked CCL1/CCR8 interaction on CCR8 single-positive cells (**A**) and CCR8/CTLA-4 double-positive cells (**B**).

GBD201 is a unique partial blocker for CTLA-4

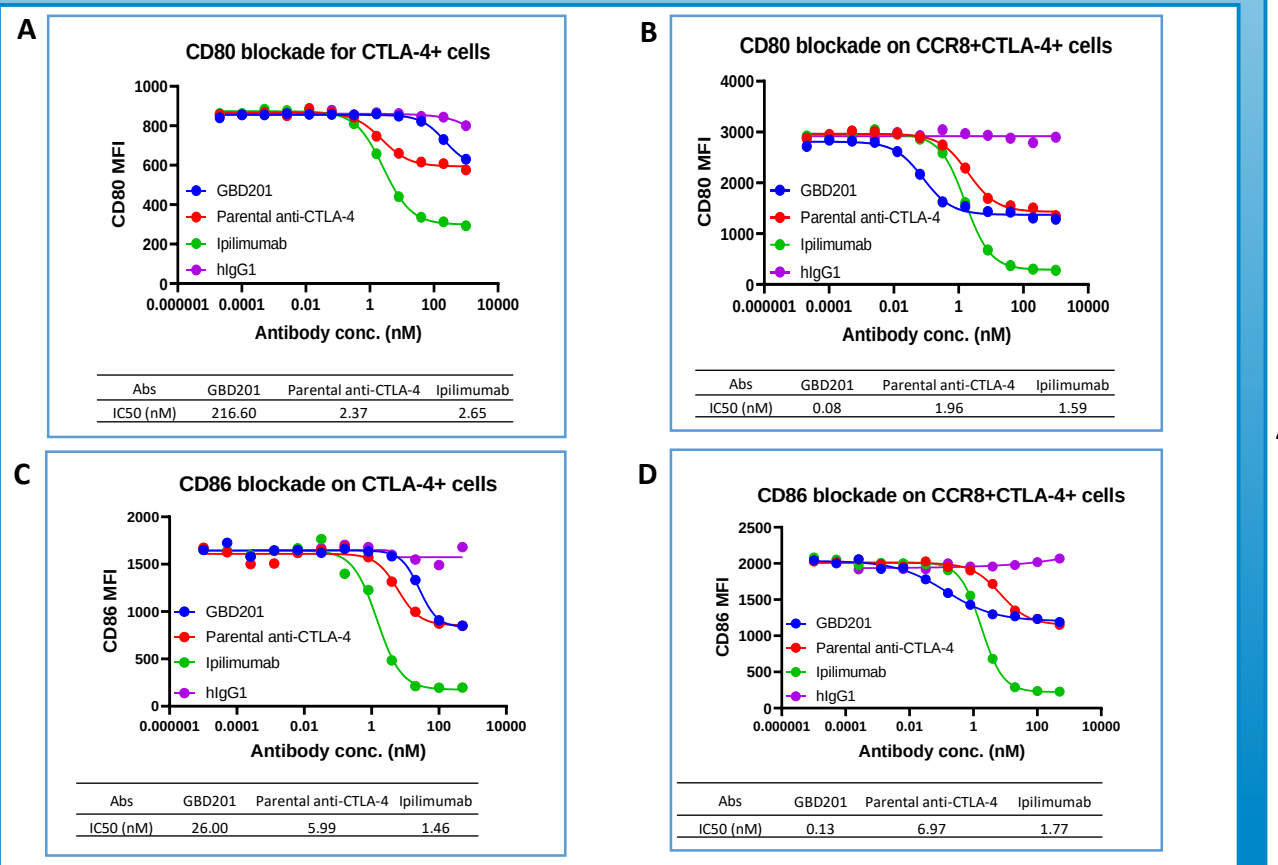
A
CD80 blockade for CTLA-4+ cells
CD80 MFI
GBD201
Parental anti-CTLA-4
Ipilimumab
hIgG1
Antibody conc. (nM)
Abs | GBD201 | Parental anti-CTLA-4 | Ipilimumab
IC50 (nM) | 216.60 | 2.37 | 2.65
B
CD80 blockade on CCR8+CTLA-4+ cells
Abs | GBD201 | Parental anti-CTLA-4 | Ipilimumab
IC50 (nM) | 0.08 | 1.96 | 1.59
C
CD86 blockade on CTLA-4+ cells
CD86 MFI
Abs | GBD201 | Parental anti-CTLA-4 | Ipilimumab
IC50 (nM) | 26.00 | 5.99 | 1.46
D
CD86 blockade on CCR8+CTLA-4+ cells
Abs | GBD201 | Parental anti-CTLA-4 | Ipilimumab
IC50 (nM) | 0.13 | 6.97 | 1.77

Figure 5. GBD201 partially blocked the interactions of CTLA-4 with its ligands (CD80 or CD86). GBD201 showed very weak blocking activity on CTLA-4 single-positive cells (**A, C**) but potent blocking activity was seen on CCR8/CTLA-4 double-positive cells (**B, D**). The partial blockade and weak binding to CTLA-4 single positive cells were intentionally designed to reduce the toxicity of CTLA-4 inhibition in the periphery.

GBD201 induced strong ADCC responses

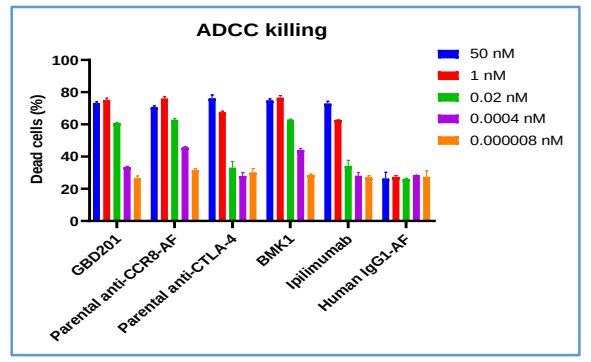
ADCC killing
Dead cells (%)
50 nM
1 nM
0.02 nM
0.0004 nM
0.000008 nM
GBD201
Parental anti-CCR8-AF
Parental anti-CTLA-4
BMK1
Ipilimumab
Human IgG1-AF

Figure 6. GBD201 induced potent ADCC killing of CCR8/CTLA-4 double-positive cells. Human PBMC were mixed with target cells (CCR8/CTLA-4 double-positive cells) at the E:T ratio of 20:1. After 2h incubation, dead cells was stained by propidium Iodide and analyzed by FACS.

In vivo characterization of GBD201 for efficacy vs. toxicity

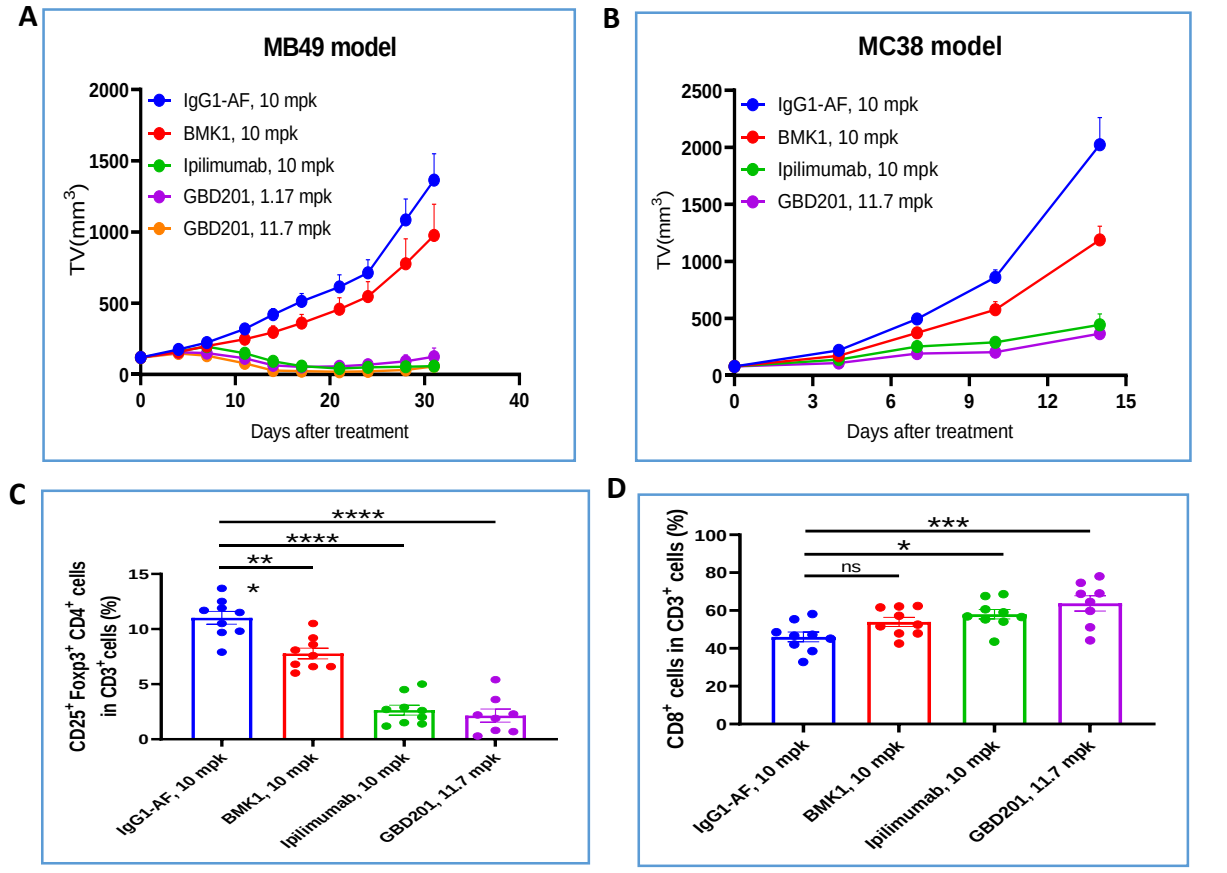
A
MB49 model
TV(mm3)
IgG1-AF, 10 mpk
BMK1, 10 mpk
Ipilimumab, 10 mpk
GBD201, 1.17 mpk
GBD201, 11.7 mpk
Days after treatment
B
MC38 model
IgG1-AF, 10 mpk
BMK1, 10 mpk
Ipilimumab, 10 mpk
GBD201, 11.7 mpk
Days after treatment
C
CD25+ Foxp3+ CD4+ cells in CD3+ cells (%)
D
CD8+ cells in CD3+ cells (%)
ns
IgG1-AF, 10 mpk
BMK1, 10 mpk
Ipilimumab, 10 mpk
GBD201, 11.7 mpk

Figure 7. GBD201 showed potent in vivo anti-tumor efficacy in syngeneic models. hCCR8/hCTLA-4 C57BL/6 mice were implanted s.c. with MB49 **(A)** or MC38 **(B)** cells and treated with Abs from 11 days (for MB49) or 7 days (for MC38) after tumor inoculation. GBD201 showed potent tumor inhibition in MB49 model (**A**) and MC38 model (**B**). The tumor infiltration lymphocytes (TILs) from MC38 model were analyzed for Treg (**C**) and CD8+T cells (**D**). Significant Treg depletion and increased CD8+ cells were observed after GBD201 treatment.
BMK1, anti-CCR8 mAb.

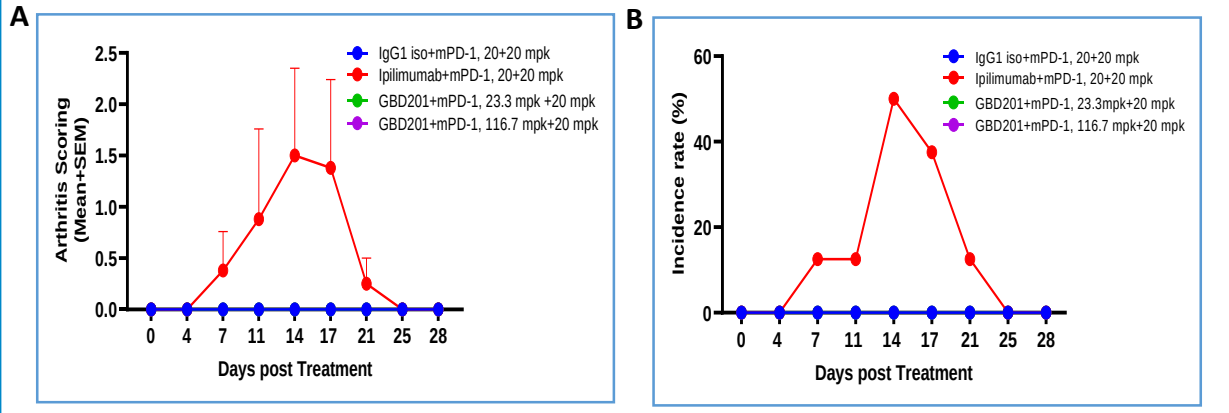
A
Arthritis Scoring (Mean+SEM)
IgG1 iso+mPD-1, 20+20 mpk
Ipilimumab+mPD-1, 20+20 mpk
GBD201+mPD-1, 23.3 mpk +20 mpk
GBD201+mPD-1, 116.7 mpk+20 mpk
Days post Treatment
B
Incidence rate (%)
Days post Treatment

Figure 8. GBD201 showed better tolerance compared with ipilimumab in the induction of arthritis. hCCR8/hCTLA4 BALB/c mice were injected with Abs i.p. on days 0, 4, 7, 11, 14 and 17. The arthritis scores for paws (**A**) and arthritis incidence rate (**B**) were evaluated. No arthritis was observed in the GBD201-treated groups (both low or high doses).

Summary

- GBD201 preferentially targets CCR8⁺CTLA-4⁺ Tregs in TME but not CTLA-4⁺ Tregs in the periphery, reducing the peripheral toxicity caused by CTLA-4 inhibition.
- GBD201 is a full blocker for CCR8, and potently inhibits CCR8/CCL1 interaction, resulting in the inhibition of Treg migration and functions.
- GBD201 has been fine tuned for the activity of CTLA-4 arm, by using a partial blocker and a unique molecular design, to balance the efficacy and safety. GBD201 showed much better safety profile compared to Ipilimumab in the arthritis induction model.
- GBD201 showed potent anti-tumor activity in mouse models. GBD201 may potentially be a next generation of CTLA-4 inhibitor by preferentially directed to TME.

Contact information: Shuhua.Han@genorbio.com